Abstract #3089: A Phase I/II study of first-in-human trial of JAB-21822 (KRAS G12C inhibitor) in advanced solid tumors

J. Li[1], J. Zhao1[1], B. S. Cao[2], J. Fang[1], X. Y. Li[3], M. Z. Wang[4], Y. Ba[5], X. Y. Li[6], Z. H. Li[7], Z. Liu[8], Y. S. Wang[9], Y. Cheng[10], C. M. Bai[11], L. Shen[5]

Beijing Cancer Hospital[1], Beijing, China; Peking University Third Hospital[2], Beijing, China; Beijing Tiantan Hospital[3], Capital Medical University[4], Beijing, China; Perking Union Medical Hospital[5], Beijing, China; Tianjin Medical University Cancer Institute & Hospital[6], Tianjin, China; The First Affiliated Hospital of Zhengzhou University[7], Zhengzhou, China; SUN YAT-SEN Memorial Hospital, SUN YAT-SEN University[8], Guangzhou, China; Beijing Chest Hospital, Capital Medical University[9], Beijing, China; West China hospital, Sichuan University[10], Chengdu, China; Jilin Cancer Hospital[11], Changchun, China

BACKGROUND

- KRAS^{G12C} mutations act as oncogenic drivers. In China, these mutations occur in approximately 4.3% of lung cancer, 2.5% of colorectal cancer (CRC), and 2.3% of biliary cancer. [1]
- JAB-21822 (Jacobio, China) is a novel, highly selective, orally bioavailable, covalent KRAS^{G12C} inhibitor
- Preclinical data have demonstrated that JAB-21822:
 - Has potent *in vitro* and *in vivo* antitumor activity
 - Has better oral bioavailability resulted in higher drug exposure compared with two leading KRAS^{G12C} inhibitors in US
 - Exhibits favorable safety profile with no risk of QT prolongation
- We present the data of the phase I portion of study (NCT05009329)

METHODS

- Key eligibility criteria
 - Advanced or metastatic solid tumor with preferable KRAS^{G12C} mutation
 - Adequate organ functions
 - Patients with advanced solid tumors who are refractory or intolerable to standard of care (SOC), or are not willing to receive SOC therapy
 - No more than 3 lines of prior therapy (specifically NSCLC expansion cohort)
 - Measurable disease according to RECIST v 1.1
 - No active brain or spinal metastases
 - ECOG 0 or 1

Figure 1. JAB-21822 Study Design*

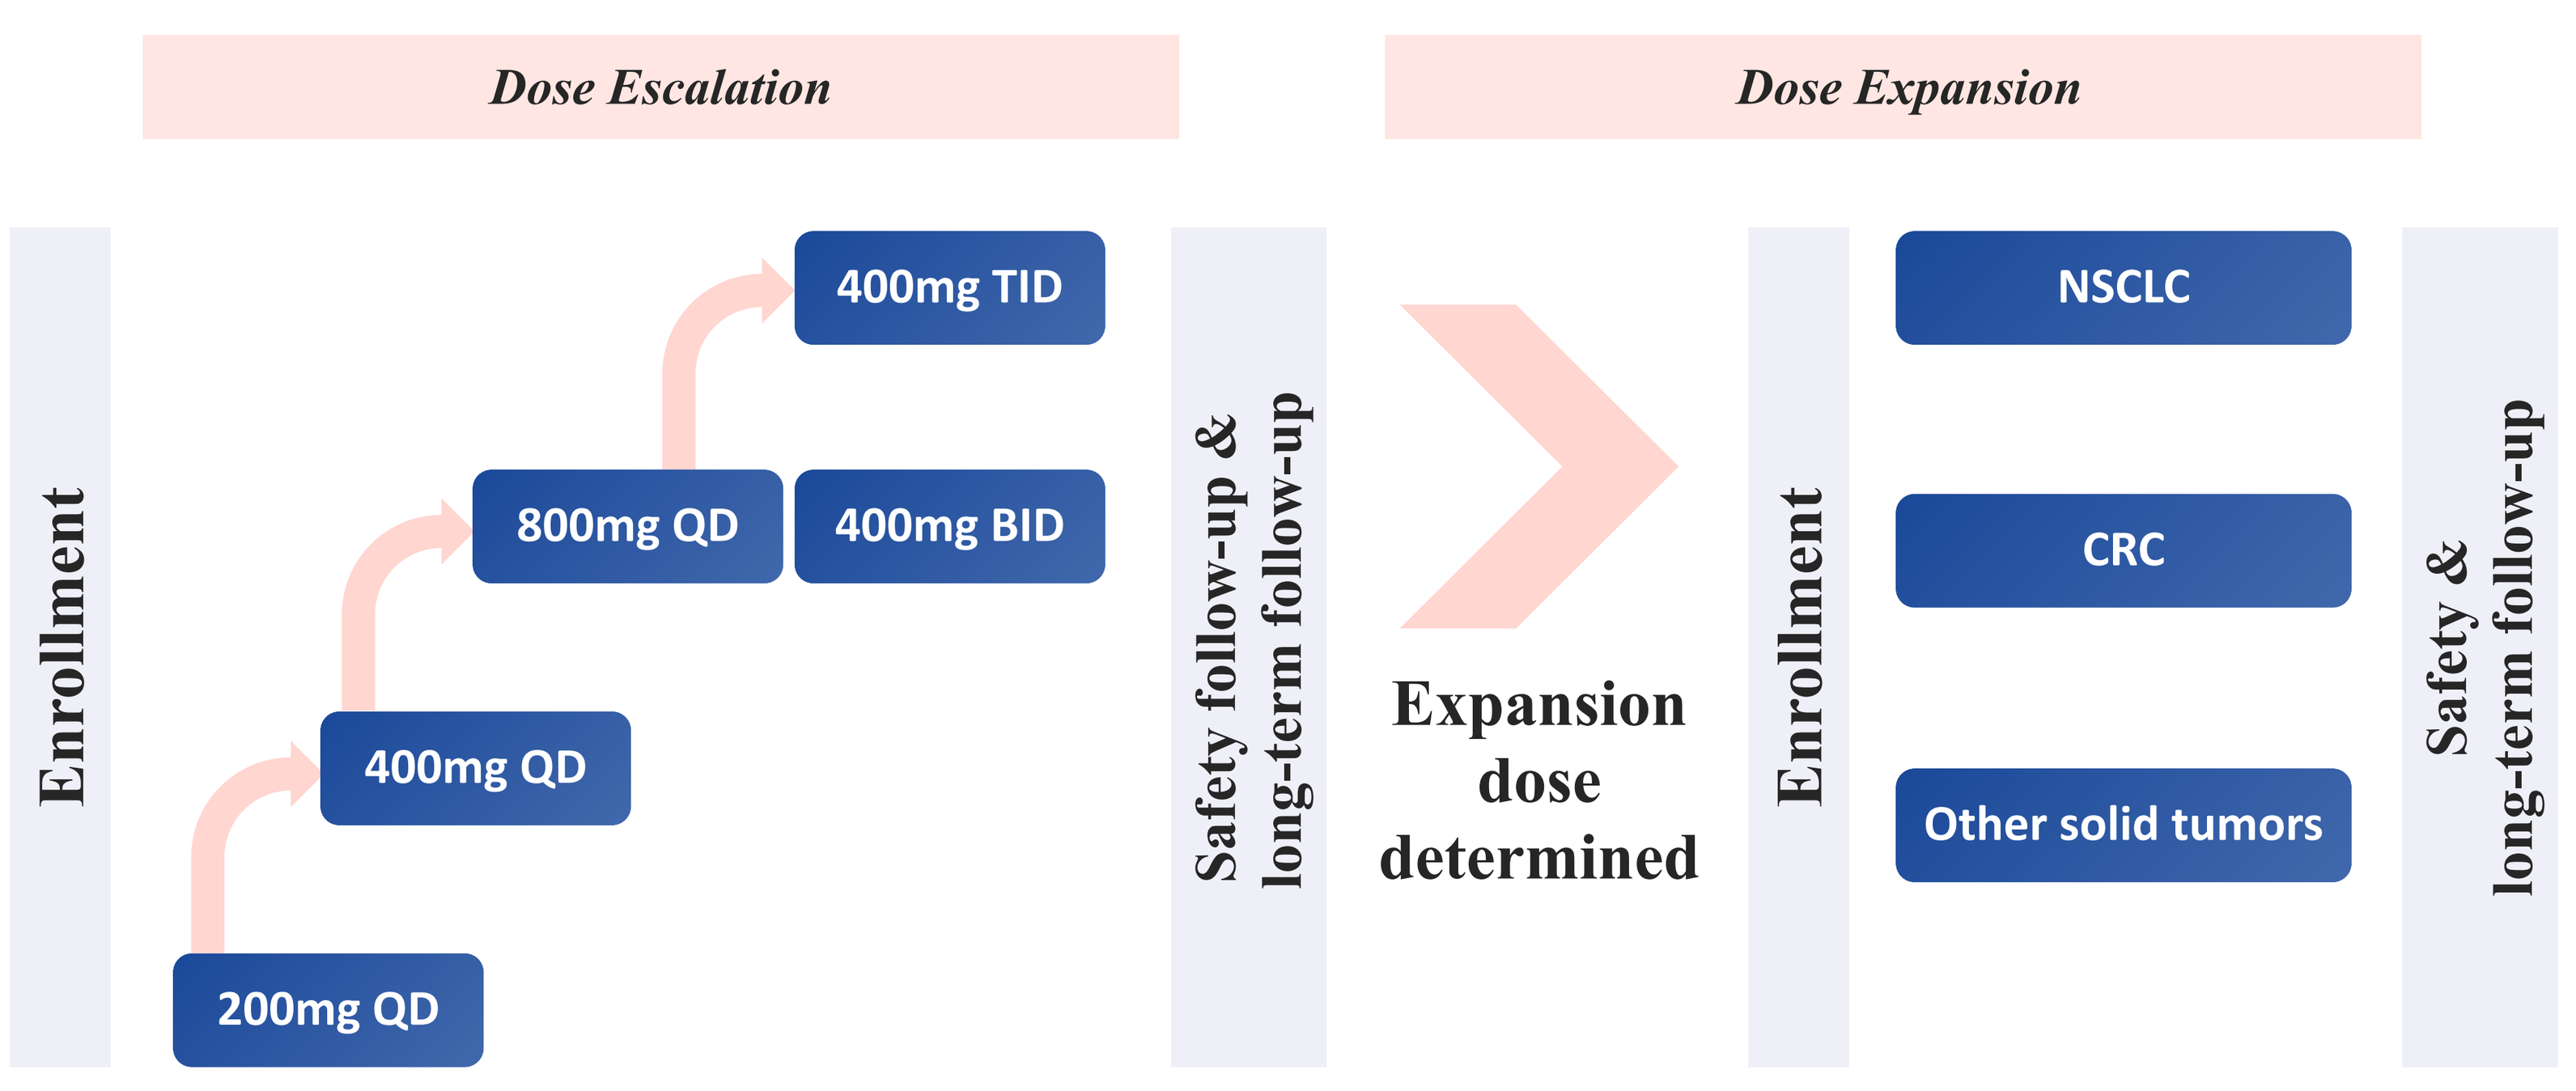
Dose Escalation
Dose Expansion
Enrollment
200mg QD
400mg QD
800mg QD
400mg BID
400mg TID
Safety follow-up & long-term follow-up
Expansion dose determined
Enrollment
NSCLC
CRC
Other solid tumors
Safety & long-term follow-up

*First-in-human, open-label; accelerated titration and 3+3 design

- Study Objectives:
 - Assess safety and tolerability of JAB-21822
 - Determine maximum tolerated dose (MTD) and recommended phase 2 dose (RP2D)
 - Characterize pharmacokinetic (PK) profile of JAB-21822 after single and multiple doses
 - Evaluate preliminary antitumor activity
- The trial is being conducted in multiple centers in China
- The date of data cutoff is April 1, 2022

Acknowledgements Insert QR code and have email here, along with any acknowledgements?

RESULTS

Table 1. Baseline Characteristics

Baseline Characteristics	All Patients (N=72)
Age, years	
median (range)	62.5 (38-85)
Sex, No. (%)	
Female	22 (30.6%)
Male	50 (69.4%)
ECOG PS, No. (%)	
0	8 (11.1%)
1	64 (88.9%)
Prior lines of systemic therapy, No. (%)	
0	5 (6.9%)
1	30 (41.7%)
2	17 (23.6%)
3	13 (18.1%)
≥4	7 (9.7%)
Brain metastases, No. (%)	
Yes	12 (16.7%)
No	59 (81.9%)
Missing	1 (1.4%)
Tumor types, No. (%)	
NSCLC	52 (72.2%)
Prior anti-PD-1/L1 therapy	
Yes	35 (67.3%)
No	17 (32.7%)
CRC	17 (23.6%)
PDAC	3 (4.2%)

Figure 2. JAB-21822 Pharmacokinetic Profile

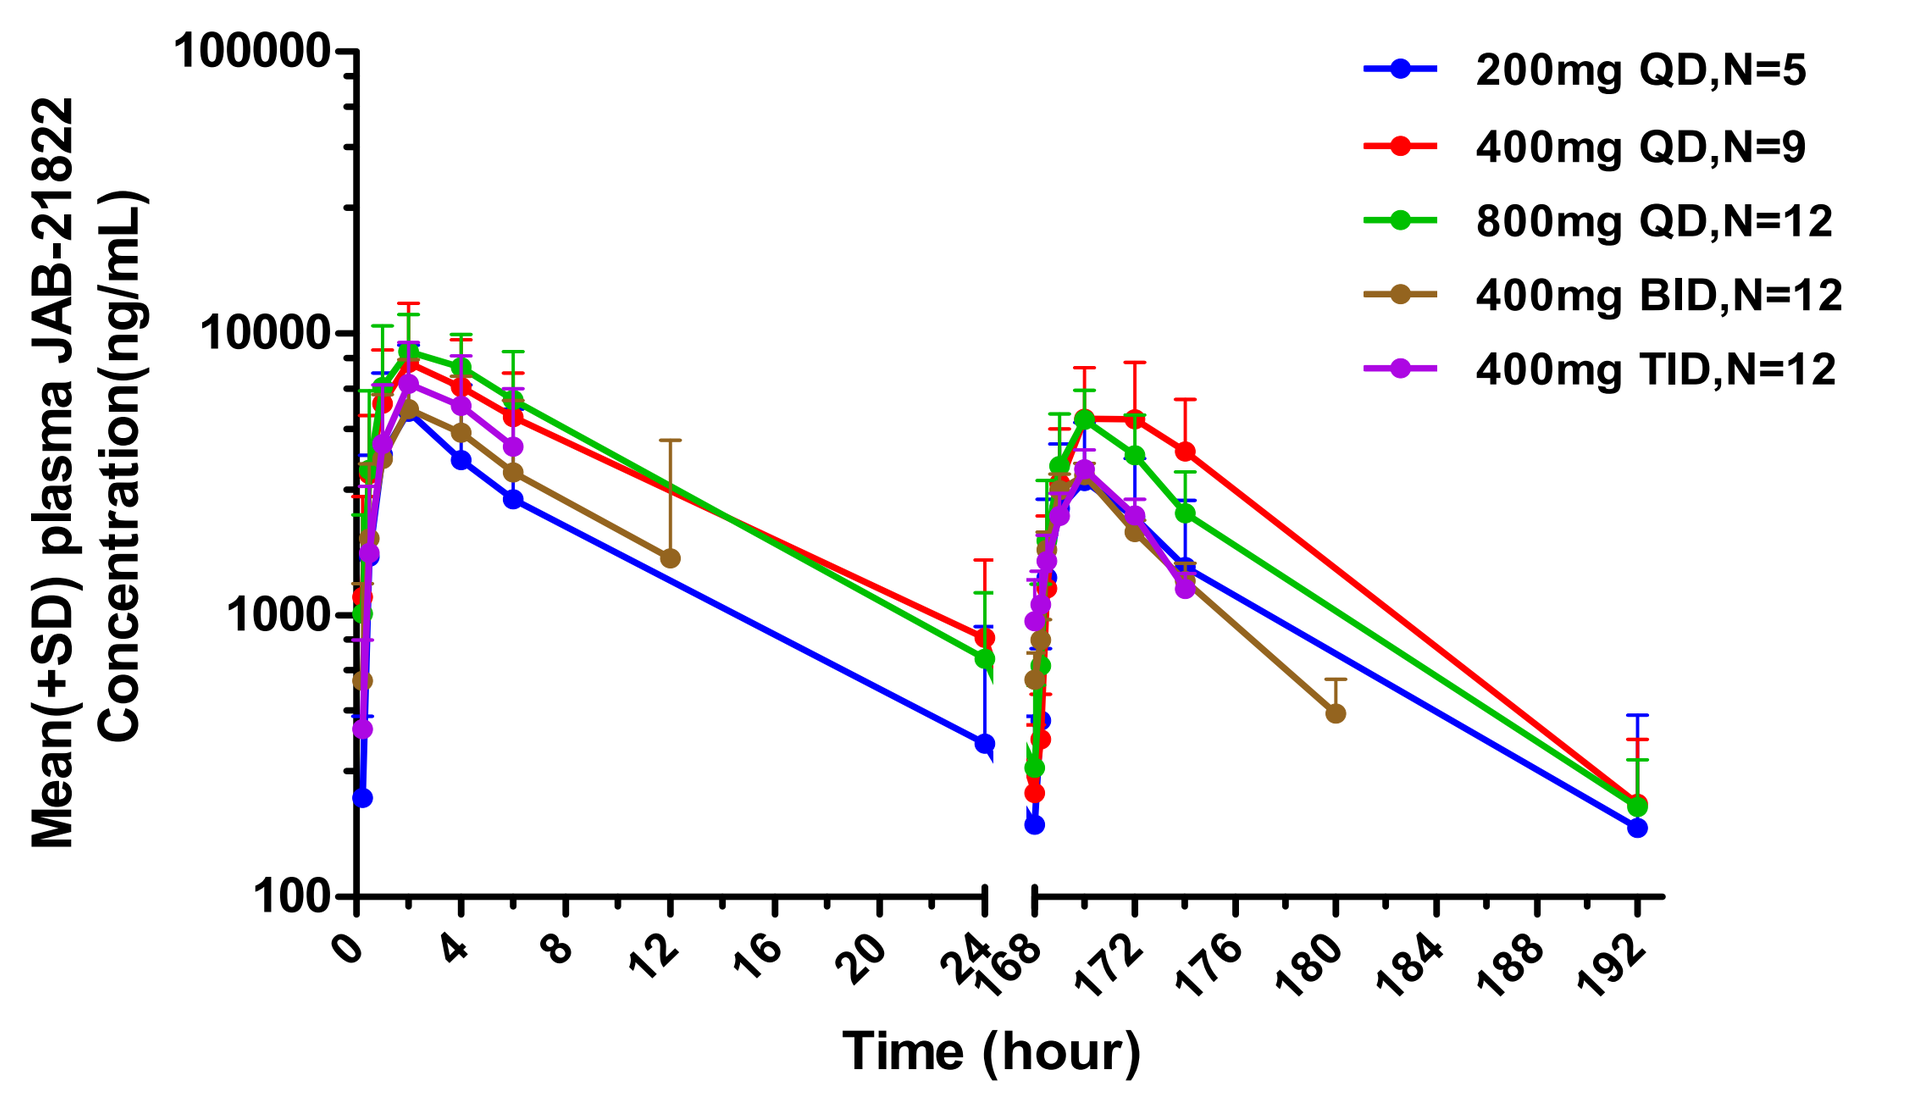
Mean(+SD) plasma JAB-21822 Concentration(ng/mL)
Time (hour)
200mg QD,N=5
400mg QD,N=9
800mg QD,N=12
400mg BID,N=12
400mg TID,N=12

PK Results

- JAB-21822 was well absorbed, with median time to reach peak plasma concentration of 2 hours
- Mean half-life was 4.99-5.54 hours

As of April 1, 2022, a total of 72 patients have received at least one dose of JAB-21822.
Two NSCLC patients in the 200 mg QD cohort did not have KRAS^{G12C} mutation.
Most patients are heavily treated with 51% having received ≥ 2 prior lines of therapy.

Table 2. Safety Summary: Treatment related adverse events (TRAE) with frequency > 5% in patients

AE	Total N=72	200 mg QD (N=5)		400 mg QD (N=18)		800 mg QD (N=22)		400 mg BID (N=12)		400 mg TID (N=15)	
	Any Grade	Grade 1-2	Grade 3-4	Grade 1-2	Grade 3-4	Grade 1-2	Grade 3-4	Grade 1-2	Grade 3-4	Grade 1-2	Grade 3-4
Blood bilirubin increased	29 (40.3%)	2 (40.0%)	0	3 (16.7%)	0	5 (22.7%)	1 (4.5%)	8 (66.7%)	0	9 (60.0%)	1 (6.7%)
Anemia	21 (29.2%)	1 (20.0%)	0	4 (22.2%)	0	5 (22.7%)	0	6 (50.0%)	0	5 (33.3%)	0
Alanine aminotransferase* increased	11 (15.3%)	0	0	3 (16.7%)	0	2 (9.1%)	0	2 (16.7%)	2 (16.7%)	1 (6.7%)	1 (6.7%)
Aspartate aminotransferase* increased	11 (15.3%)	0	0	3 (16.7%)	0	1 (4.5%)	0	3 (25.0%)	2 (16.7%)	1 (6.7%)	1 (6.7%)
Proteinuria	8 (11.1%)	0	0	1 (5.6%)	0	1 (4.5%)	0	4 (33.3%)	0	2 (13.3%)	0
White blood cell count decreased	7 (9.7%)	0	0	1 (5.6%)	0	1 (4.5%)	0	3 (25.0%)	0	1 (6.7%)	1 (6.7%)
Neutrophil count decreased	5 (6.9%)	0	0	1 (5.6%)	0	1 (4.5%)	0	0	2 (16.7%)	0	1 (6.7%)
Rash	5 (6.9%)	0	0	0	0	2 (9.1%)	0	1 (8.3%)	0	2 (13.3%)	0
Vomiting	5 (6.9%)	0	0	3 (16.7%)	0	1 (4.5%)	0	0	0	1 (6.7%)	0
Diarrhea	4 (5.6%)	0	0	0	0	2 (9.1%)	0	2 (16.7%)	0	0	0
Hyponatremia	4 (5.6%)	0	0	1 (5.6%)	0	1 (4.5%)	0	2 (16.7%)	0	0	0

- Most common AE (frequency ≥ 10%) amongst all cohorts: Blood bilirubin increased, anemia, alanine aminotransferase and aspartate aminotransferase increased, proteinuria
- Majority of TRAEs are grades 1-2
- Lower Grade 3-4 TRAEs in QD cohort comparing with BID or TID cohort
- No TRAEs led to discontinuation
- No Grade 5 TRAEs were observed
- 3 patients (4.2%) experienced treatment-related SAEs

Table 3: Objective Response Rate in Patients with KRAS^{G12C} solid tumors

	200 MG QD (N=2)	400 MG QD (N=9)	800 MG QD (N=11)	400 MG BID (N=12)	400 MG TID (N=15)	All Patients (N=49*)
Best Overall Response No. (%)						
Complete Response (CR)	0	0	0	0	0	0
Partial Response (PR)	0	5 (55.6%)	5 (45.5%)	3 (25.0%)	7 (46.7%)	20 (40.8%)
Stable Disease (SD)	2 (100.0%)	4 (44.4%)	6 (54.5%)	7 (58.3%)	6 (40.0%)	25 (51.0%)
Progressive Disease (PD)	0	0	0	2 (16.7%)	2 (13.3%)	4 (8.2%)
Objective Response Rate (ORR)	0	5 (55.6%)	5 (45.5%)	3 (25.0%)	7 (46.7%)	20 (40.8%)
Disease Control Rate (DCR)	2 (100.0%)	9 (100%)	11 (100%)	10 (83.3%)	13 (86.7%)	45 (91.8%)

*Efficacy evaluable patient is defined as having completed at least one post-treatment assessment. At data cutoff, 19 patients have not reached 1st post-treatment tumor assessment.

- All solid tumors:
 - ORR 40.8% (20/49)
 - DCR 91.8% (45/49)
- KRAS^{G12C} mutant NSCLC: (Figure 3 and 4)

RESULTS - KRAS^{G12C} NSCLC

Figure 3: Patients with KRAS^{G12C} mutant NSCLC

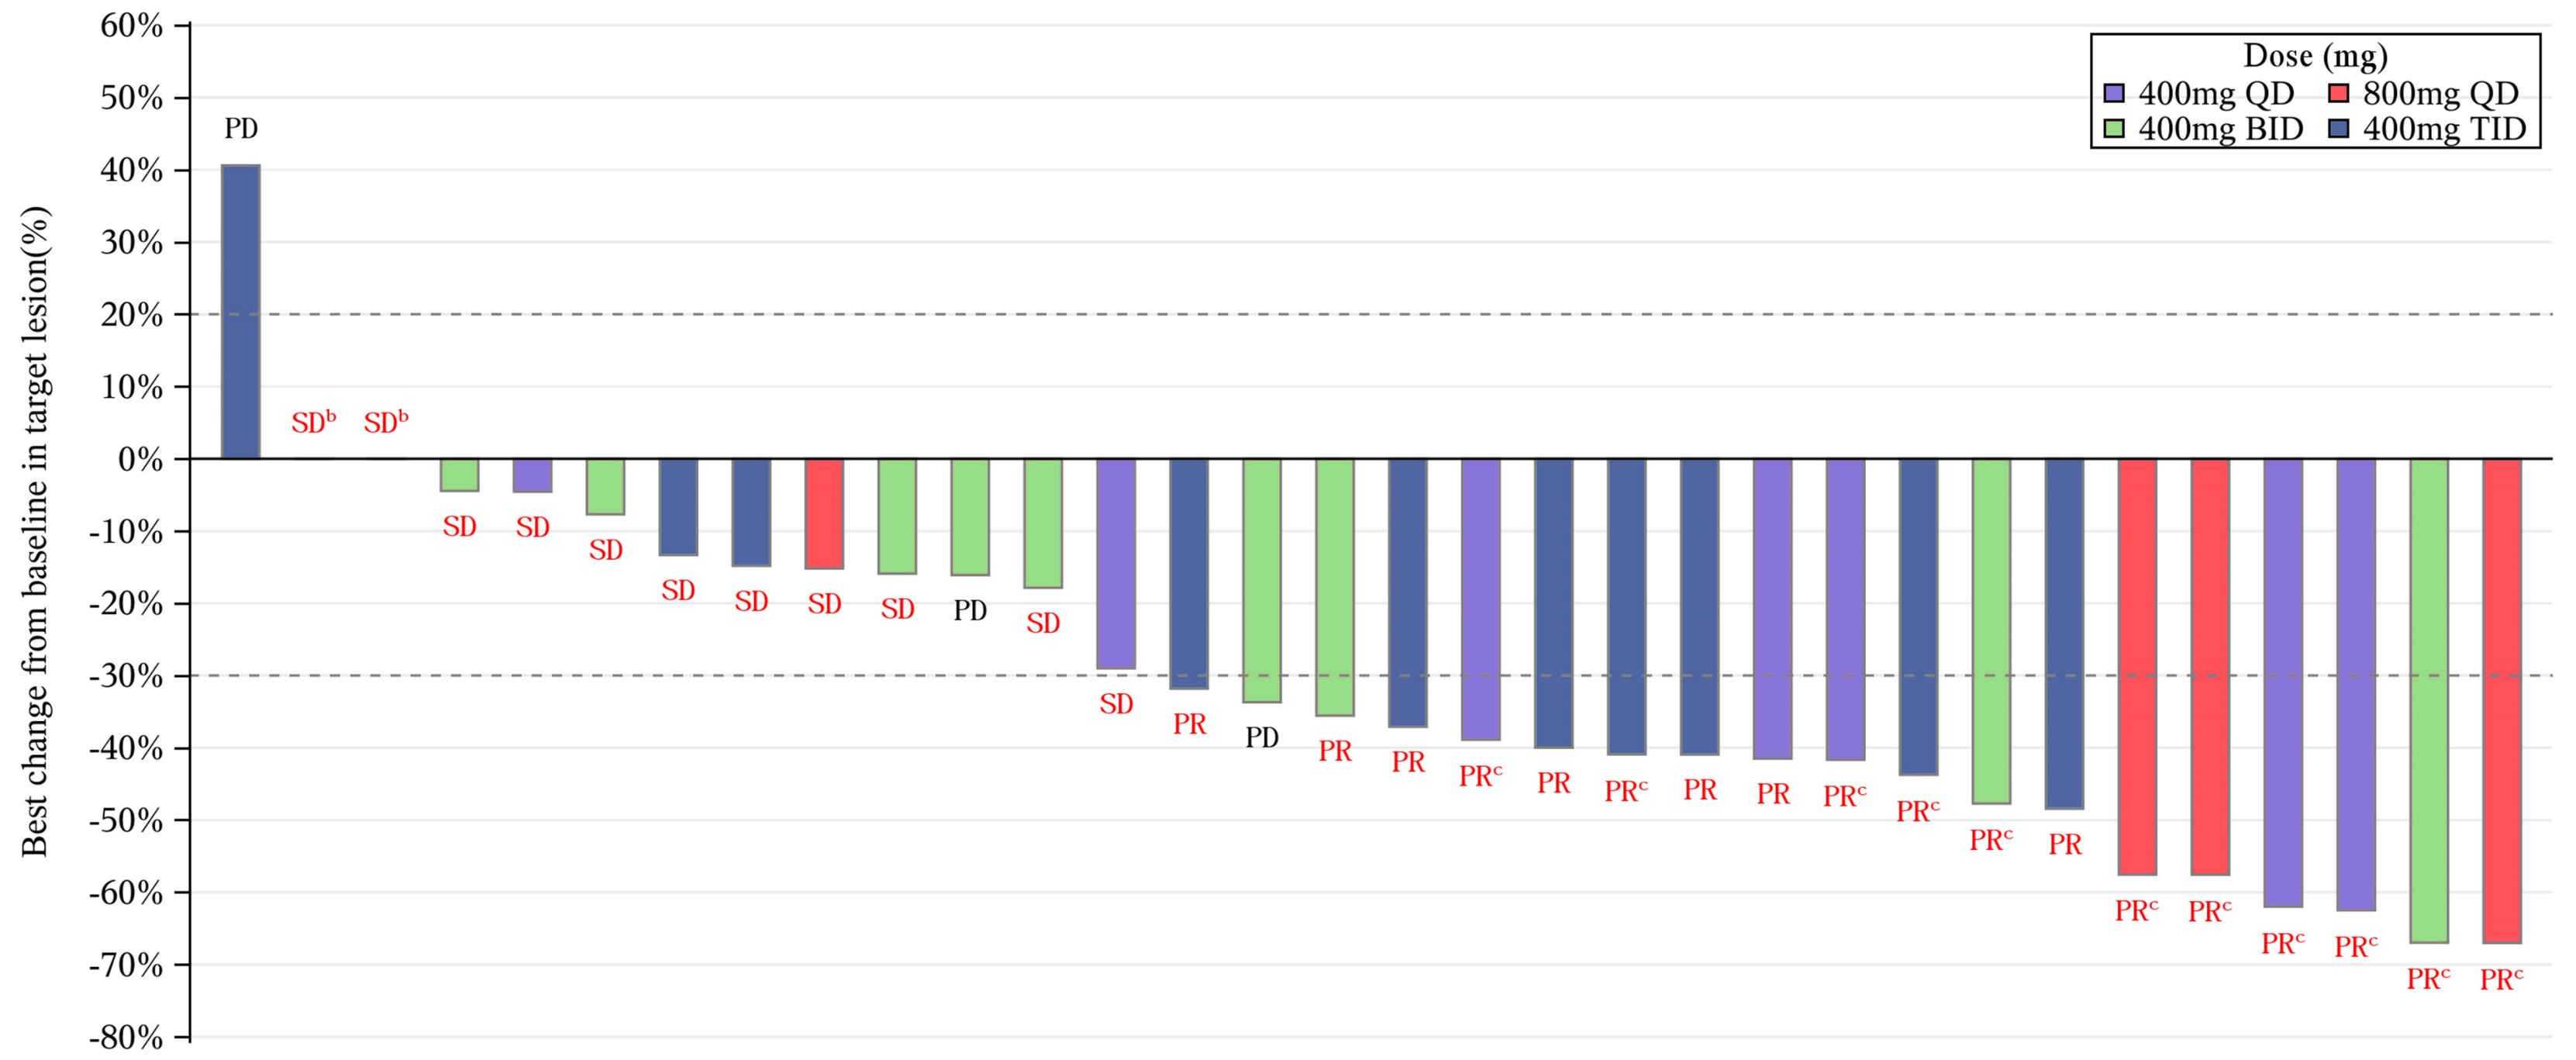
Best change from baseline in target lesion(%)
Dose (mg)
400mg QD
800mg QD
400mg BID
400mg TID

b: one patient 800 mg QD and one patient 400 mg BID; c: confirmed PR

- **ORR 56.3% (18/32); DCR 90.6% (29/32)**
- QD COHORT (included 400mg & 800mg QD):
 - **ORR for 400 mg and 800 mg QD cohorts is 66.7% (8/12)**
 - **DCR for QD dosing 400 mg and 800 mg 100% (12/12)**

Figure 4: Treatment duration: Patients with KRAS^{G12C} mutant NSCLC

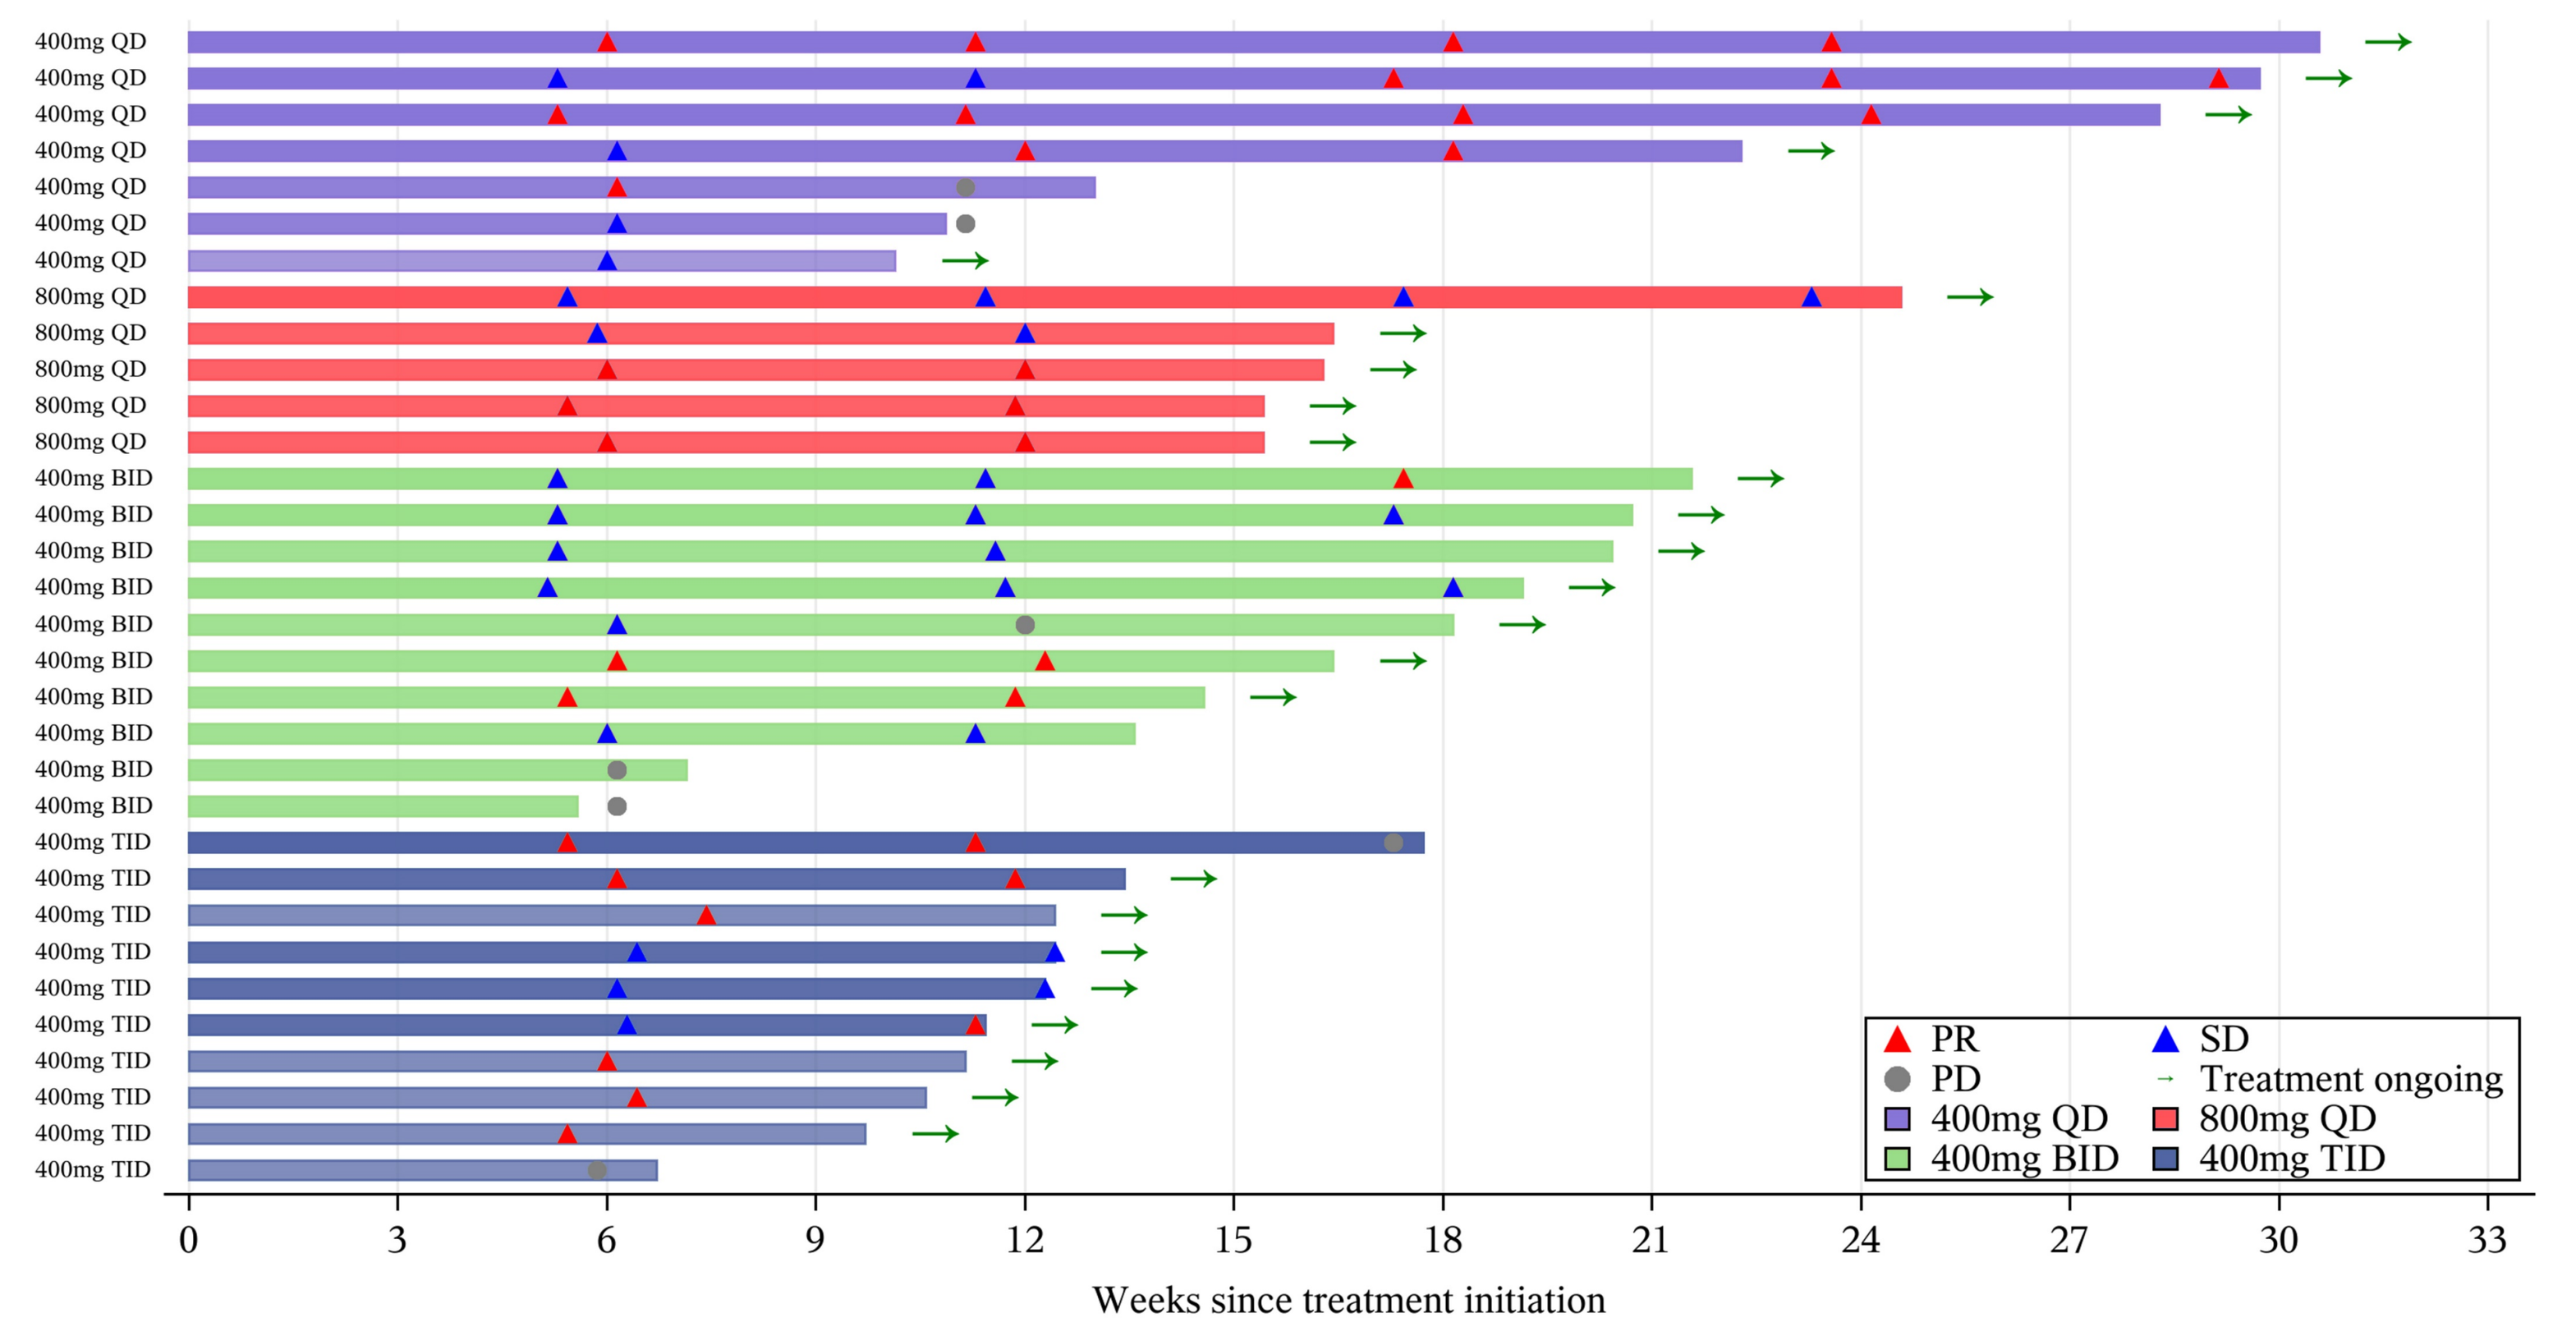
Weeks since treatment initiation
PR
SD
PD
Treatment ongoing
400mg QD
800mg QD
400mg BID
400mg TID

CONCLUSION

- JAB-21822 is well tolerated with no DLTs in the dose escalation phase
- In KRAS^{G12C} mutant NSCLC : Overall, ORR 56.3% (18/32), DCR 90.6% (29/32); in QD cohort, ORR 66.7% (8/12) and DCR 100% (12/12).
- Preliminary efficacy results demonstrate promising clinical activity of single-agent JAB-21822 in patients with KRAS^{G12C} mutant NSCLC
- Subject recruitment is ongoing, and trial remains open to enrollment

REFERENCES

1.Loong HH, et al. Transl Lung Cancer Res. 2020;9(5):1759-1769.